Tislelizumab With or Without Capecitabine Continuation in Gastric or Gastro-oesophageal Junction Cancer: RATIONALE-305 Post Hoc Analysis

2100P

Markus Moehler,[1]* Rui-Hua Xu,[2] Filippo Pietrantonio,[3] Sun Young Rha,[4] Ken Kato,[5] María Alsina Maqueda,[6] Mohammed Najeeb Al Hallak,[7] Elizabeth C Smyth,[8] Tianshu Liu,[9] Tobias Arkenau,[10] Josep Tabernero,[11] Yaling Xu,[12] Xuan Kong,[12] Sheng Xu,[12] David Wang[13]

[1]Johannes Gutenberg-University Clinic, Mainz, Germany; [2]Sun Yat-sen University Cancer Center, State Key Laboratory of Oncology in South China, Collaborative Innovation Center of Cancer Medicine, Guangzhou, China; [3]National Cancer Institute Fondazione IRCCS of Milan, Milan, Italy; [4]Severance Hospital, Yonsei University Health System, Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Korea; [5]Department of Head and Neck, Esophageal Medical Oncology, National Cancer Center Hospital, Tokyo, Japan; [6]Hospital Universitario de Navarra, Pamplona, Spain; [7]Karmanos Cancer Institute, Wayne State University, Detroit, MI, USA; [8]Oxford University NHS Foundation Trust, Oxford, UK; [9]Zhongshan Hospital, Fudan University, Shanghai, China; [10]Department of Medical Oncology, The Christie NHS Foundation Trust/Division of Cancer Sciences, University of Manchester, Manchester, UK; [11]Department of Oncology, Vall d'Hebron University Hospital and Institute of Oncology (VHIO), Barcelona, Spain; [12]BeOne Medicines, Ltd., Shanghai, China; [13]University of Michigan, Ann Arbor, MI, USA. *Lead and presenting author.

CONCLUSIONS

- In advanced gastric or gastro-oesophageal junction cancer (GC/GEJC), patients who completed six cycles of tislelizumab plus CAPOX (capecitabine plus oxaliplatin) or placebo plus CAPOX and continued capecitabine had substantially longer survival vs those who discontinued capecitabine (median overall survival [OS]: 21.0 months vs 10.2 months with tislelizumab; 18.1 months vs 12.3 months with placebo)
- Patients who received tislelizumab with capecitabine continuation demonstrated significantly longer OS compared with those who received placebo with capecitabine continuation (21.0 vs 18.1 months; hazard ratio [HR]=0.78; nominal *P*=.0071)
- Tislelizumab with capecitabine continuation treatment showed significant improvements in OS, progression-free survival (PFS), and objective response rate (ORR) vs placebo with capecitabine continuation
- Continuation of capecitabine with tislelizumab did not result in any new safety signals, making this a suitable option after tislelizumab plus CAPOX in patients with locally advanced or metastatic GC/GEJC
- Investigators had a strong preference to administer capecitabine continuation when patients were eligible
- Among patients who received CAPOX without capecitabine continuation, PFS and ORR were numerically higher with tislelizumab vs placebo, but not statistically significant, and no OS advantage was observed

INTRODUCTION

- Standard systemic therapy for GC/GEJC typically includes fluoropyrimidines (5-fluorouracil [5-FU] or capecitabine), platinum agents (oxaliplatin or cisplatin), and, in the modern era, immune checkpoint inhibitors (ICIs) for advanced disease[1-3]
- Tislelizumab, an anti–programmed cell death protein-1 (PD-1) monoclonal antibody, demonstrated improved OS when combined with chemotherapy in the phase 3 RATIONALE-305 trial (NCT03777657), which evaluated tislelizumab or placebo with CAPOX or cisplatin plus 5-FU[4]
- Preclinical studies suggest that oxaliplatin-based regimens may synergise more effectively with PD-1 inhibitors like tislelizumab compared with other platinum agents through immunogenic cell death mechanisms[5]
- No high-level evidence has been generated for continuation therapy with ICIs alone or with capecitabine after CAPOX induction for advanced GC/GEJC, although small retrospective studies have suggested potential benefits of capecitabine maintenance.[6-8] Continuation strategies may have significant implications for patients' quality of life, highlighting the importance of evaluating patient values in maintenance therapy decision-making[9]
- Here we present the first post hoc analysis of a phase 3 trial evaluating the efficacy and safety of tislelizumab with capecitabine continuation or placebo with capecitabine continuation after induction treatment with six cycles of tislelizumab plus CAPOX or placebo plus CAPOX in patients with human epidermal growth factor receptor 2 (HER2)-negative, advanced GC/GEJC

METHODS

- RATIONALE-305 was a randomised, double-blind, placebo-controlled phase 3 trial comparing tislelizumab plus chemotherapy with chemotherapy alone in treatment-naive patients with locally advanced or metastatic GC/GEJC[4,10]
- Eligible patients with HER2-negative, unresectable locally advanced or metastatic GC/GEJC were randomised 1:1 to receive tislelizumab or placebo in combination with CAPOX or cisplatin plus 5-FU for up to six cycles. Capecitabine continuation with tislelizumab or placebo was at the investigator's discretion (**Figure 1**)
- This analysis included patients who received tislelizumab plus CAPOX or placebo plus CAPOX and who were alive and remained on study after completing six cycles of chemotherapy
- A sensitivity analysis was conducted, excluding patients who had experienced disease progression after six cycles, to assess the robustness of findings
- The primary endpoint was OS in patients with programmed death-ligand 1 (PD-L1)–positive Tumor Area Positivity (TAP) score ≥5% and in all patients regardless of PD-L1 expression
- All efficacy endpoints (OS, PFS, ORR) were measured from the time of enrollment; all *P*-values are nominal

Figure 1. Study Design

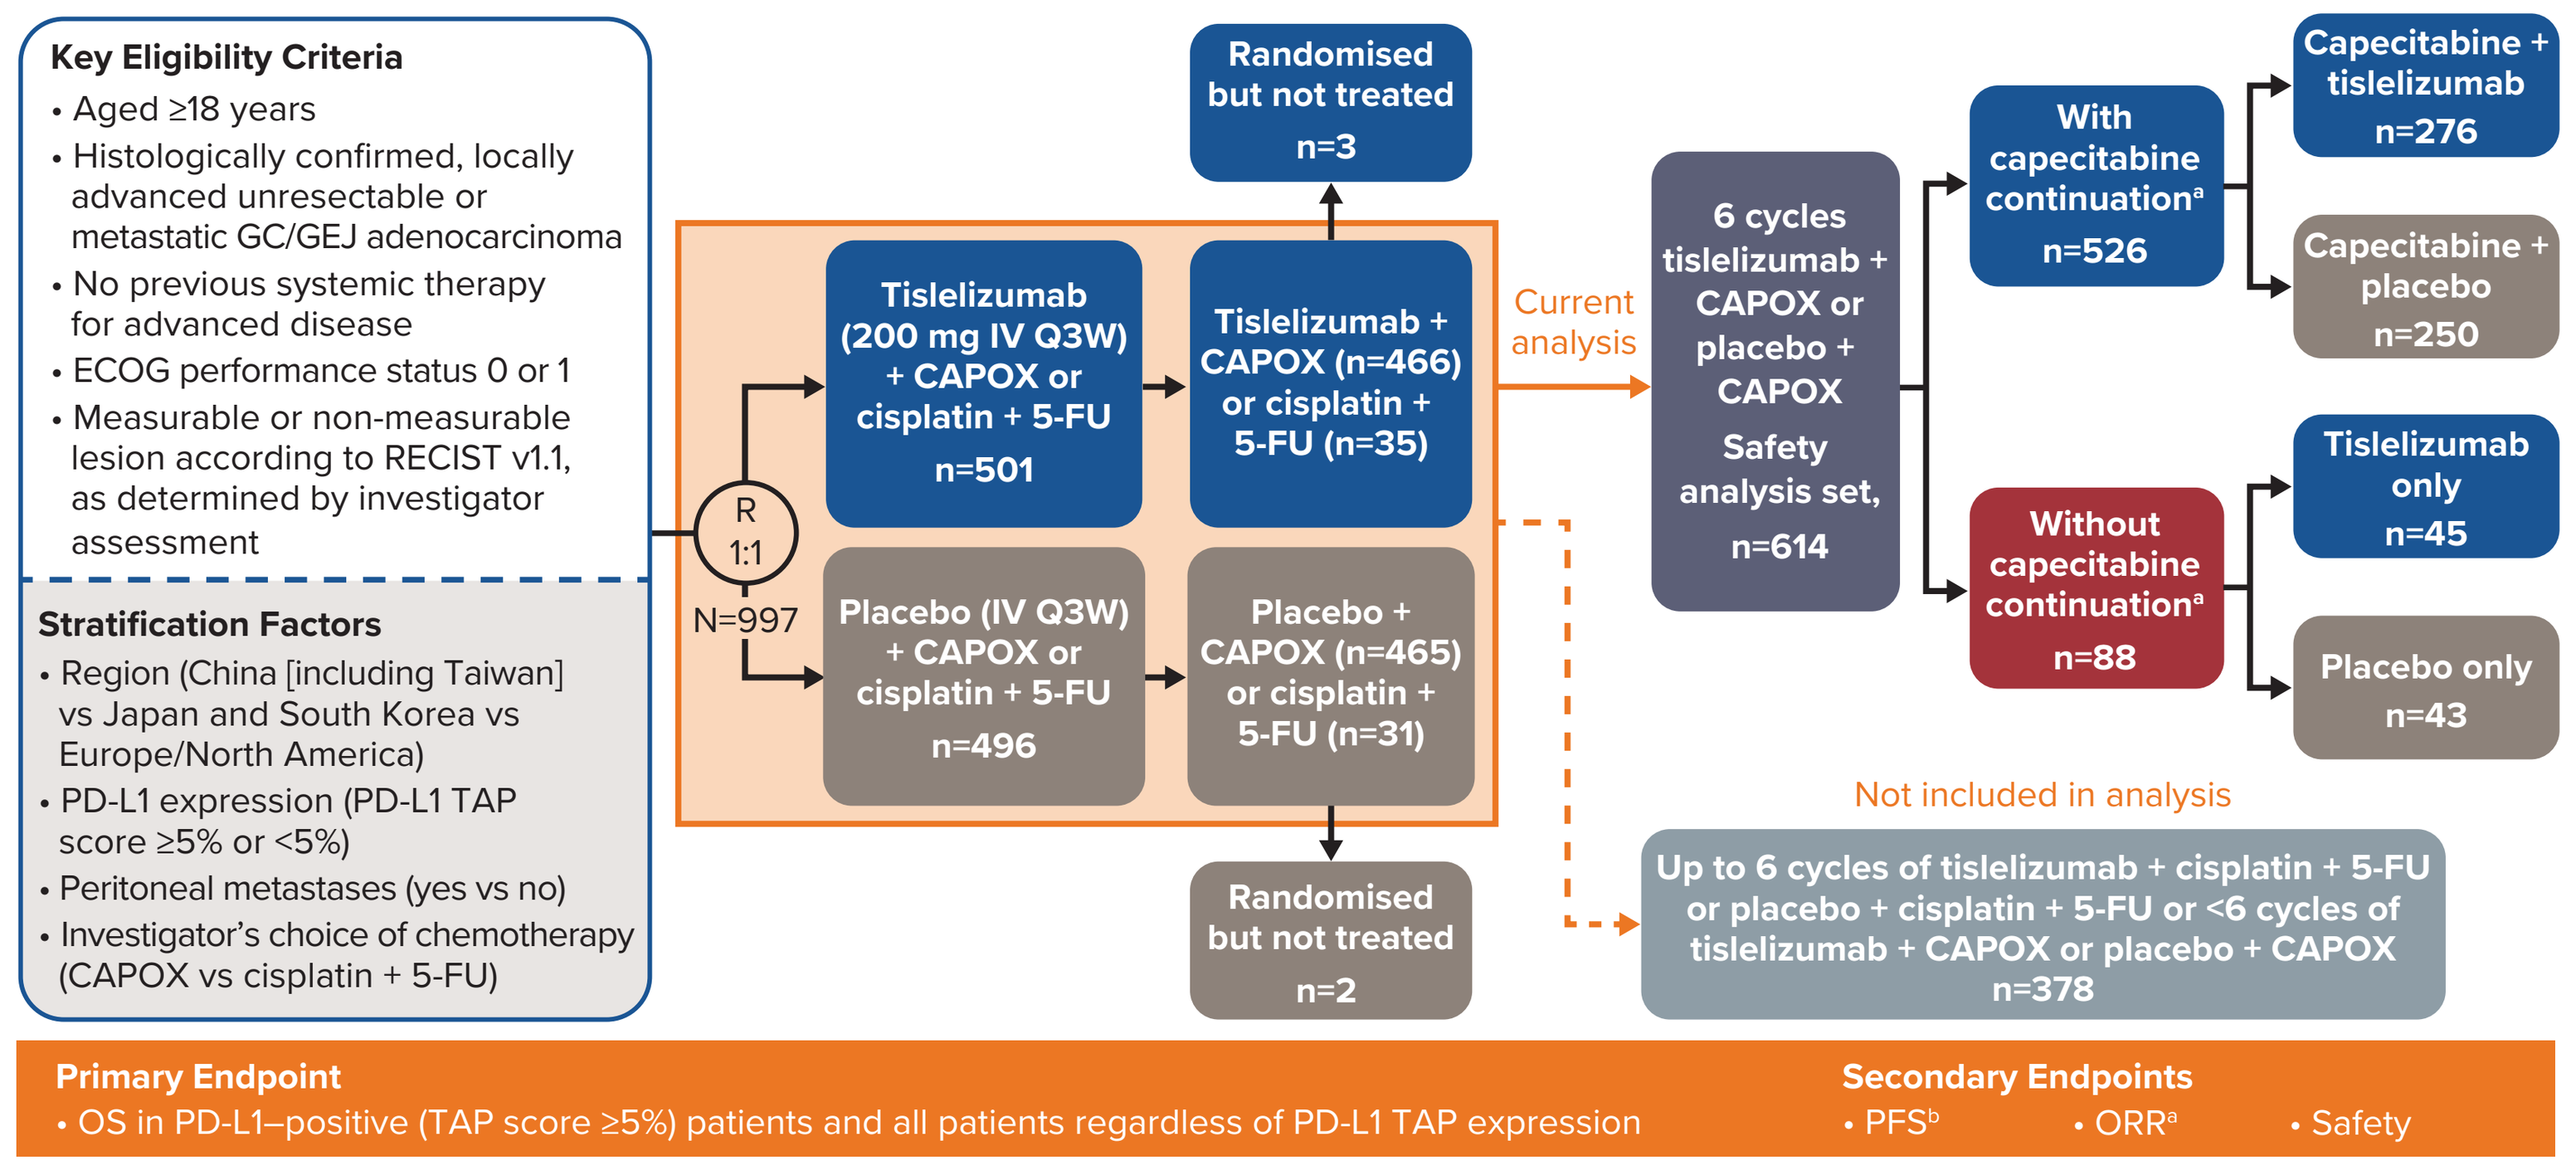
Key Eligibility Criteria
• Aged ≥18 years
• Histologically confirmed, locally advanced unresectable or metastatic GC/GEJ adenocarcinoma
• No previous systemic therapy for advanced disease
• ECOG performance status 0 or 1
• Measurable or non-measurable lesion according to RECIST v1.1, as determined by investigator assessment
Stratification Factors
• Region (China [including Taiwan] vs Japan and South Korea vs Europe/North America)
• PD-L1 expression (PD-L1 TAP score ≥5% or <5%)
• Peritoneal metastases (yes vs no)
• Investigator's choice of chemotherapy (CAPOX vs cisplatin + 5-FU)
R 1:1
N=997
Tislelizumab (200 mg IV Q3W) + CAPOX or cisplatin + 5-FU n=501
Placebo (IV Q3W) + CAPOX or cisplatin + 5-FU n=496
Randomised but not treated n=3
Tislelizumab + CAPOX (n=466) or cisplatin + 5-FU (n=35)
Placebo + CAPOX (n=465) or cisplatin + 5-FU (n=31)
Randomised but not treated n=2
Current analysis
6 cycles tislelizumab + CAPOX or placebo + CAPOX
Safety analysis set, n=614
With capecitabine continuation[a] n=526
Without capecitabine continuation[a] n=88
Capecitabine + tislelizumab n=276
Capecitabine + placebo n=250
Tislelizumab only n=45
Placebo only n=43
Not included in analysis
Up to 6 cycles of tislelizumab + cisplatin + 5-FU or placebo + cisplatin + 5-FU or <6 cycles of tislelizumab + CAPOX or placebo + CAPOX n=378
Primary Endpoint
• OS in PD-L1–positive (TAP score ≥5%) patients and all patients regardless of PD-L1 TAP expression
Secondary Endpoints
• PFS[b] • ORR[a] • Safety

[a]Continuation of capecitabine was at the discretion of the investigator and per their clinical judgement. [b]Investigator assessed per RECIST v1.1.
Abbreviations: ECOG, Eastern Cooperative Oncology Group; IV, intravenous; Q3W, every 3 weeks; RECIST, Response Evaluation Criteria in Solid Tumors.

RESULTS

Patient Disposition and Baseline Characteristics

- A total of 997 patients were randomised, of whom 931 (93.4%) received tislelizumab plus CAPOX or placebo plus CAPOX; 614 (61.6%) received all six cycles of tislelizumab plus CAPOX or placebo plus CAPOX and were eligible for continuation therapy
- Among these 614 eligible patients, 526 (85.7%) received tislelizumab plus capecitabine or placebo plus capecitabine as continuation therapy (tislelizumab: n=276, placebo: n=250)
- Nearly all patients eligible for continuation therapy (608/614; 99.0%) had metastatic disease at baseline
- Median study follow-up time was 46.4 months (95% confidence interval [CI]: 44.2, 49.5) for tislelizumab and 47.3 months (95% CI: 43.7, 51.9) for placebo in the capecitabine continuation group, and 51.5 months (95% CI: 45.2, not evaluated [NE]) for tislelizumab and 53.2 months (95% CI: 45.2, NE) for placebo in the group without capecitabine continuation

RESULTS (CONT.)

- Baseline demographics and disease characteristics were similar across treatment arms (**Table 1**)

Table 1. Patient Baseline Demographics and Characteristics (Safety Analysis Set)

	With Capecitabine Continuation		Without Capecitabine Continuation	
	Tislelizumab (n=276)	Placebo (n=250)	Tislelizumab (n=45)	Placebo (n=43)
Median age, years	60.0	61.0	57.0	61.0
<65, n (%)	195 (70.7)	155 (62.0)	36 (80.0)	27 (62.8)
Sex, n (%)				
Male	188 (68.1)	174 (69.6)	30 (66.7)	36 (83.7)
Female	88 (31.9)	76 (30.4)	15 (33.3)	7 (16.3)
Region, n (%)				
East Asia[a]	232 (84.1)	206 (82.4)	33 (73.3)	33 (76.7)
Rest of world[b]	44 (15.9)	44 (17.6)	12 (26.7)	10 (23.3)
ECOG performance status, n (%)				
0	96 (34.8)	71 (28.4)	16 (35.6)	15 (34.9)
1	180 (65.2)	179 (71.6)	29 (64.4)	28 (65.1)
Primary location, n (%)				
Gastro-oesophageal junction	42 (15.2)	33 (13.2)	9 (20.0)	12 (27.9)
Stomach	234 (84.8)	217 (86.8)	36 (80.0)	31 (72.1)
PD-L1 TAP expression, n (%)				
<5%	121 (43.8)	120 (48.0)	18 (40.0)	17 (39.5)
≥5%	155 (56.2)	130 (52.0)	27 (60.0)	26 (60.5)

[a]Includes China, Taiwan, Japan, and South Korea. [b]Includes the United States and Europe.

Efficacy

- Median treatment duration was longer in patients receiving capecitabine continuation than in those without continuation (tislelizumab: 10.0 vs 4.4 months; placebo: 9.6 vs 4.4 months)
- At data cutoff (August 27, 2024), patients who received tislelizumab with capecitabine continuation showed significant improvements in efficacy outcomes compared with those who received placebo with capecitabine continuation (**Table 2**):
 - Patients who received tislelizumab with capecitabine continuation demonstrated significantly longer OS compared with those who received placebo with capecitabine continuation (**Figure 2**)
 - Small numbers of patients in the group without capecitabine continuation limit the interpretation
 - PFS was significantly improved with tislelizumab vs placebo in patients with capecitabine continuation and numerically improved with tislelizumab vs placebo in patients without capecitabine continuation (**Figure 3**)
 - ORR was higher with tislelizumab vs placebo, both in patients with capecitabine continuation and without capecitabine continuation

Table 2. Efficacy Outcomes (Safety Analysis Set)

	With Capecitabine Continuation		Without Capecitabine Continuation	
	Tislelizumab (n=276)	Placebo (n=250)	Tislelizumab (n=45)	Placebo (n=43)
Median OS, months (95% CI)	21.0 (18.6, 24.1)	18.1 (16.4, 20.3)	10.2 (7.2, 14.1)	12.3 (11.1, 15.2)
HR (95% CI)	0.78 (0.65, 0.95)		1.24 (0.80, 1.92)	
Median PFS[a], months (95% CI)	10.1 (9.0, 12.9)	9.5 (8.3, 9.8)	4.7 (4.2, 5.5)	4.3 (4.1, 4.5)
HR (95% CI)	0.79 (0.65, 0.96)		0.82 (0.52, 1.32)	
ORR[a], % (95% CI)	71.4 (65.7, 76.6)	66.8 (60.6, 72.6)	57.8 (42.2, 72.3)	48.8 (33.3, 64.5)

[a]PFS and ORR were derived from tumour assessments by investigators.

Figure 2. Kaplan–Meier Plot of OS With Capecitabine Continuation (Safety Analysis Set)

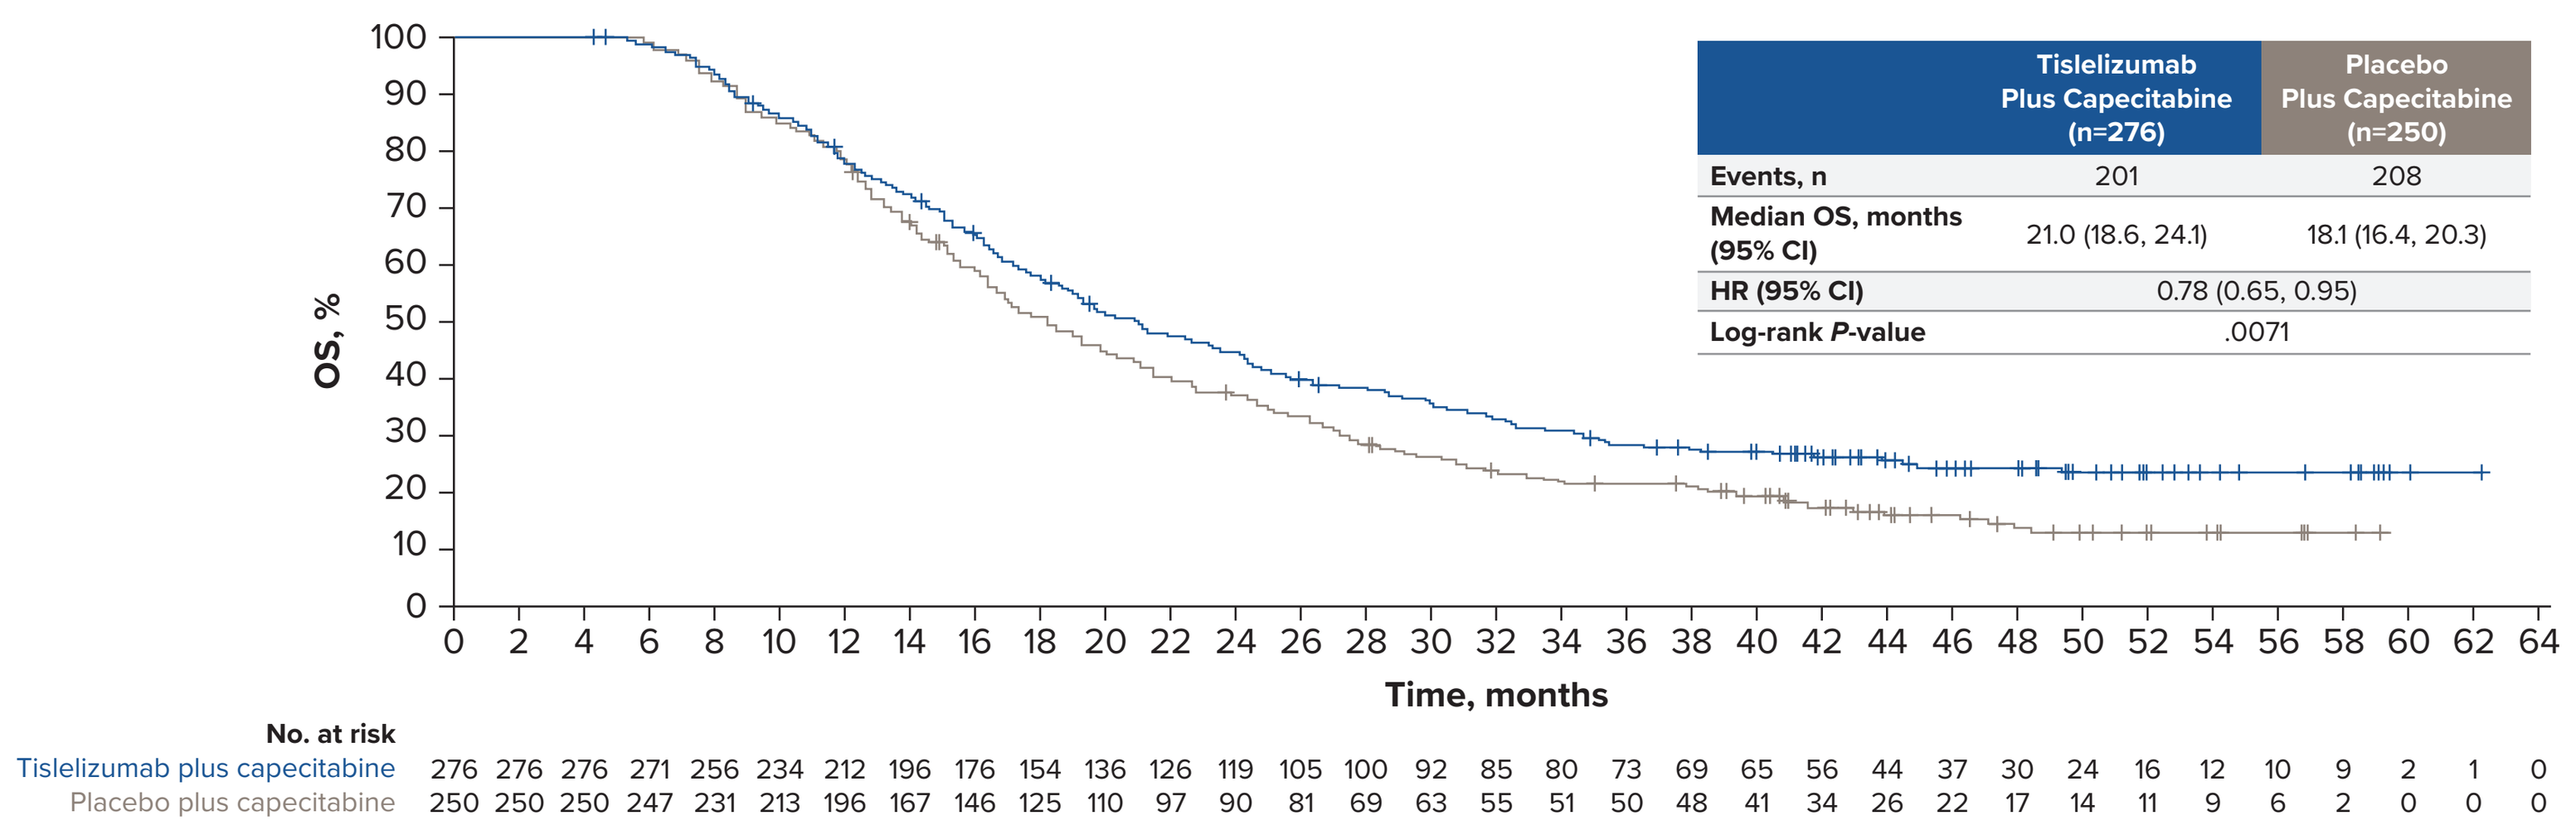
OS, %
100
90
80
70
60
50
40
30
20
10
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44 46 48 50 52 54 56 58 60 62 64
Time, months
Tislelizumab Plus Capecitabine (n=276) | Placebo Plus Capecitabine (n=250)
Events, n 201 208
Median OS, months (95% CI) 21.0 (18.6, 24.1) 18.1 (16.4, 20.3)
HR (95% CI) 0.78 (0.65, 0.95)
Log-rank P-value .0071
No. at risk
Tislelizumab plus capecitabine 276 276 276 271 256 234 212 196 176 154 136 126 119 105 100 92 85 80 73 69 65 56 44 37 30 24 16 12 10 9 2 1 0
Placebo plus capecitabine 250 250 250 247 231 213 196 167 146 125 110 97 90 81 69 63 55 51 50 48 41 34 26 22 17 14 11 9 6 2 0 0 0

Figure 3. Kaplan–Meier Plot of PFS With Capecitabine Continuation (Safety Analysis Set)

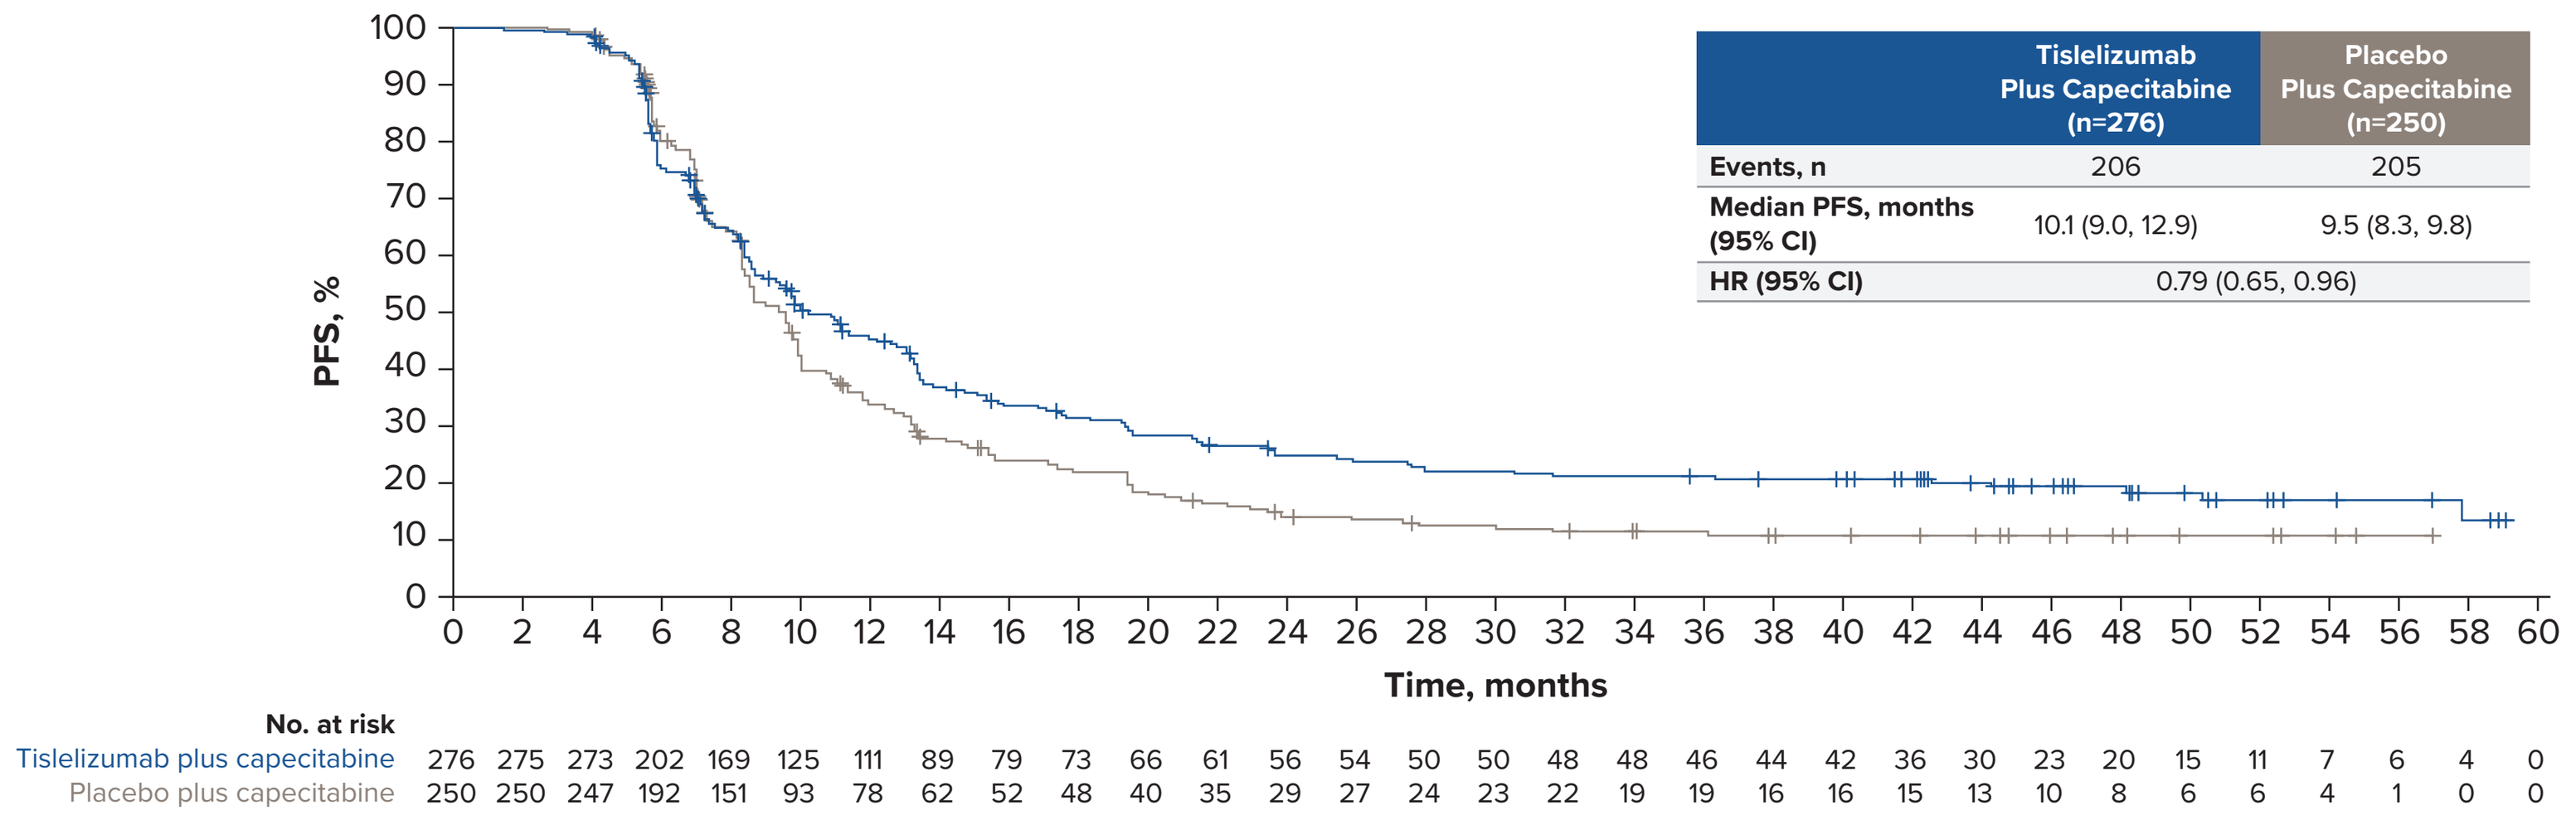
PFS, %
100
90
80
70
60
50
40
30
20
10
0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44 46 48 50 52 54 56 58 60
Time, months
Tislelizumab Plus Capecitabine (n=276) | Placebo Plus Capecitabine (n=250)
Events, n 206 205
Median PFS, months (95% CI) 10.1 (9.0, 12.9) 9.5 (8.3, 9.8)
HR (95% CI) 0.79 (0.65, 0.96)
No. at risk
Tislelizumab plus capecitabine 276 275 273 202 169 125 111 89 79 73 66 61 56 54 50 50 48 48 46 44 42 36 30 23 20 15 11 7 6 4 0
Placebo plus capecitabine 250 250 247 192 151 93 78 62 52 48 40 35 29 27 24 23 22 19 19 16 16 15 13 10 8 6 6 4 1 0 0

- In PD-L1 subgroup analyses with capecitabine continuation, greater treatment benefit was observed among patients with higher tumour PD-L1 expression (PD-L1 TAP score ≥1% and ≥5%) receiving tislelizumab vs placebo (**Table 3**)
- OS results were consistent across all prespecified subgroups (**Figure 4**)

Table 3. Efficacy Outcomes by PD-L1 Expression TAP Score Level (With Capecitabine Continuation, Safety Analysis Set)

	TAP Score ≥1% Subgroup		TAP Score ≥5% Subgroup	
	Tislelizumab (n=238)	Placebo (n=225)	Tislelizumab (n=155)	Placebo (n=130)
Median OS, months (95% CI)	21.2 (18.1, 24.4)	18.1 (16.4, 20.3)	23.5 (18.4, 27.1)	18.1 (15.5, 20.0)
HR (95% CI)	0.77 (0.63, 0.95)		0.72 (0.55, 0.94)	
Median PFS[a], months (95% CI)	10.0 (8.6, 12.9)	9.5 (8.4, 9.9)	11.1 (9.3, 14.1)	9.4 (8.3, 9.9)
HR (95% CI)	0.80 (0.65, 0.98)		0.75 (0.58, 0.99)	
ORR[a], % (95% CI)	73.1 (67.0, 78.6)	68.4 (61.9, 74.5)	76.8 (69.3, 83.2)	70.8 (62.2, 78.4)

[a]PFS and ORR were derived from tumour assessments by investigators.

Figure 4. Forest Plot of OS With Capecitabine Continuation (Safety Analysis Set)

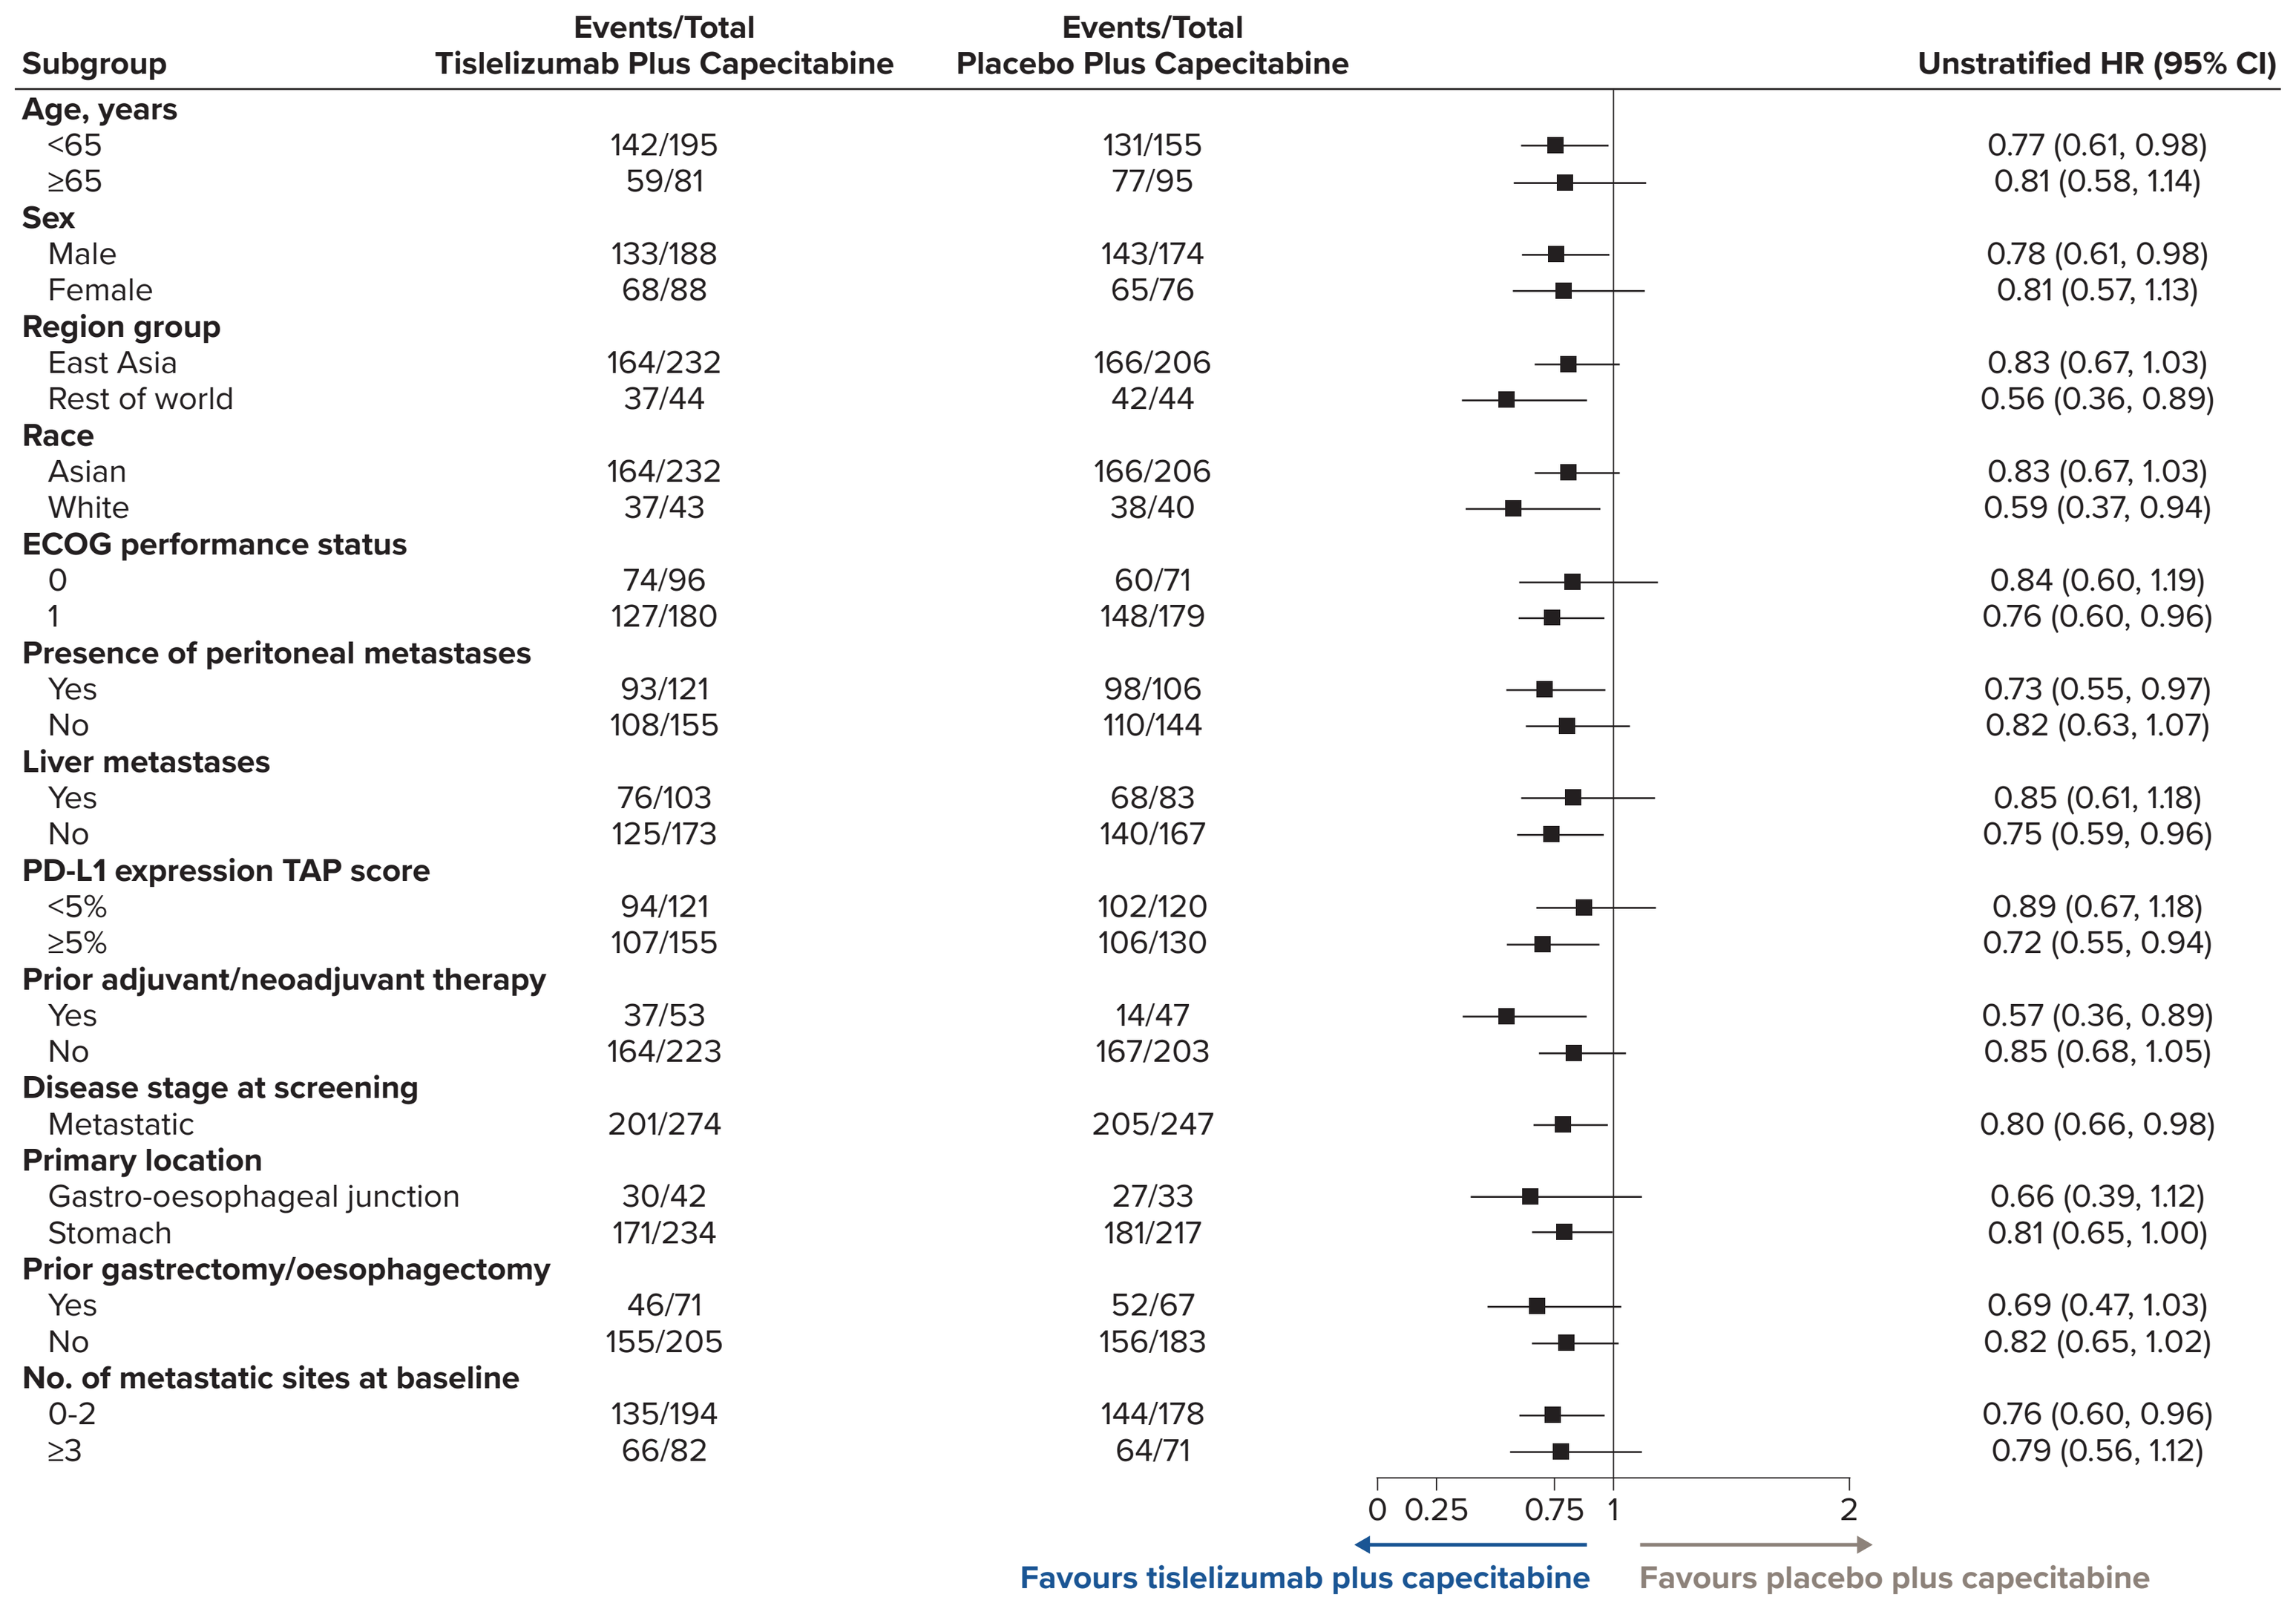
Subgroup | Events/Total Tislelizumab Plus Capecitabine | Events/Total Placebo Plus Capecitabine | Unstratified HR (95% CI)
Age, years
<65 142/195 131/155 0.77 (0.61, 0.98)
≥65 59/81 77/95 0.81 (0.58, 1.14)
Sex
Male 133/188 143/174 0.78 (0.61, 0.98)
Female 68/88 65/76 0.81 (0.57, 1.13)
Region group
East Asia 164/232 166/206 0.83 (0.67, 1.03)
Rest of world 37/44 42/44 0.56 (0.36, 0.89)
Race
Asian 164/232 166/206 0.83 (0.67, 1.03)
White 37/43 38/40 0.59 (0.37, 0.94)
ECOG performance status
0 74/96 60/71 0.84 (0.60, 1.19)
1 127/180 148/179 0.76 (0.60, 0.96)
Presence of peritoneal metastases
Yes 93/121 98/106 0.73 (0.55, 0.97)
No 108/155 110/144 0.82 (0.63, 1.07)
Liver metastases
Yes 76/103 68/83 0.85 (0.61, 1.18)
No 125/173 140/167 0.75 (0.59, 0.96)
PD-L1 expression TAP score
<5% 94/121 102/120 0.89 (0.67, 1.18)
≥5% 107/155 106/130 0.72 (0.55, 0.94)
Prior adjuvant/neoadjuvant therapy
Yes 37/53 14/47 0.57 (0.36, 0.89)
No 164/223 167/203 0.85 (0.68, 1.05)
Disease stage at screening
Metastatic 201/274 205/247 0.80 (0.66, 0.98)
Primary location
Gastro-oesophageal junction 30/42 27/33 0.66 (0.39, 1.12)
Stomach 171/234 181/217 0.81 (0.65, 1.00)
Prior gastrectomy/oesophagectomy
Yes 46/71 52/67 0.69 (0.47, 1.03)
No 155/205 156/183 0.82 (0.65, 1.02)
No. of metastatic sites at baseline
0-2 135/194 144/178 0.76 (0.60, 0.96)
≥3 66/82 64/71 0.79 (0.56, 1.12)
0 0.25 0.75 1 2
Favours tislelizumab plus capecitabine
Favours placebo plus capecitabine

- Sensitivity analysis of patients without disease progression after six cycles (tislelizumab: n=267, placebo: n=240) confirmed the treatment benefit observed in the main analysis (data not shown)

Safety

- Nearly all patients experienced at least one treatment-emergent adverse event (TEAE) across treatment groups, with treatment-related adverse events (TRAEs) reported in most patients (**Table 4**)
- Serious TRAEs were more frequent in the tislelizumab-containing arms, both with capecitabine continuation and without capecitabine continuation
- Grade ≥3 TRAEs occurred at similar rates in patients receiving tislelizumab with capecitabine continuation vs tislelizumab alone and at higher rates in patients receiving placebo with capecitabine continuation vs placebo alone
- TRAEs leading to treatment discontinuation were more frequent with tislelizumab than placebo, both with capecitabine continuation and without capecitabine continuation
- TRAEs leading to dose modifications occurred at similar rates across treatment arms, and the overall safety profile was consistent with known profiles of the individual treatment components

Table 4. Overall Safety Summary (Safety Analysis Set)

	With Capecitabine Continuation		Without Capecitabine Continuation	
n (%)	Tislelizumab (n=276)	Placebo (n=250)	Tislelizumab (n=45)	Placebo (n=43)
Patients with ≥1 TEAE	275 (99.6)	249 (99.6)	45 (100.0)	43 (100.0)
TRAE for any treatment component	272 (98.6)	247 (98.8)	45 (100.0)	43 (100.0)
Serious TRAEs for any treatment component	56 (20.3)	36 (14.4)	11 (24.4)	2 (4.7)
Grade ≥3 TRAEs for any treatment component	152 (55.1)	133 (53.2)	27 (60.0)	17 (39.5)
TRAEs leading to any treatment discontinuation	45 (16.3)	19 (7.6)	8 (17.8)	0
TRAEs leading to dose modification of any treatment component	227 (82.2)	206 (82.4)	34 (75.6)	34 (79.1)
TRAEs leading to death	4 (1.4)	2 (0.8)	1 (2.2)	0

REFERENCES

1. Lordick F, et al. *Ann Oncol*. 2022;33:1005-1020.
2. Kono K, et al. *Gastric Cancer*. 2020;23:565-578.
3. Grivas P, et al. *Target Oncol*. 2019;14:505-525.
4. Qiu M, et al. *BMJ*. 2024;385:e078876.
5. Liu P, et al. *Oncoimmunology*. 2022;11:2093518.
6. Oyan B, et al. *J Clin Oncol*. 2014;32(Suppl 15):e15003.
7. Qiu MZ, et al. *Tumor Biol*. 2014;35:4369-4375.
8. Moehler M, et al. *J Clin Oncol*. 2021;39:966-977.
9. Theis S, et al. *Front Oncol*. 2025;15:1588721.
10. Cruz-Correa M, et al. *Curr Med Res Opin*. 2025;41:1007-1016.

DISCLOSURES

MM: Consulting or advisory roles for Bayer, MSD, Merck Serono, Amgen, Taiho, Pfizer, Roche, Lilly, Servier, BeOne Medicines, Ltd., Bristol Myers Squibb, AstraZeneca, Astellas, Dragonfly, and Novartis; Honoraria from Amgen, Roche/Genentech, Merck Serono, MSD Oncology, Bristol Myers Squibb, AstraZeneca/MedImmune, Servier, Pierre Fabre, Sanofi, Transcenta, Daiichi Sankyo, Astellas, and Nordic; Grant or research funding from Amgen, Leap Therapeutics, Merck Serono, and MSD; Other renumeration from AIO, Amgen, Merck Serono, Roche, Bayer, ASCO, German Cancer Society, MSD, ESMO, BeOne Medicines, Ltd., and EORTC.

ACKNOWLEDGEMENTS

The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centres. We would also like to acknowledge John Wu of BeOne Medicines for his assistance in data analysis. This study was sponsored by BeOne Medicines, Ltd. Medical writing support was provided by Corinne Fourie, PhD, and Lauren Van Wassenhove, PhD, of Parexel, and supported by BeOne Medicines.

Presented at ESMO, October 17-21, 2025, Berlin, Germany